1647P

Clinical Validity of Plasma DNA Testing to Identify *BRCA*-mutated (*BRCA+*) Patients in the MAGNITUDE Study

Gerhardt Attard,[1*] Usha Singh,[2] Yaji Xu,[2] Lesley Farrington,[2] Katherine Bell,[3] Karen Urtishak,[3] Won Kim,[4] Jenny Zhang,[3] Sarah Parsons,[2] Kate (Yueyi) Li,[5] Melissa Schwenk,[5] Wei Li,[5] Songbai Wang[2]

[1]University College London, London, United Kingdom; [2]Janssen Research & Development, LLC, Raritan, NJ, USA; [3]Janssen Research & Development, LLC, Spring House, PA, USA; [4]Janssen Research & Development, LLC, Los Angeles, CA, USA; [5]Foundation Medicine, Inc., Cambridge, MA, USA.

*Presenting author

Key Takeaway

In patients with mCRPC considering treatment with NIRA+AAP, *BRCA* mutations can be effectively identified using the non-invasive plasma-based F1LCDx assay or the tissue-based F1CDx assay to help guide treatment decisions

Conclusions

These analyses demonstrate the clinical validity of F1LCDx in identifying *BRCA+* patients with mCRPC for NIRA+AAP

– In the above study, satisfactory concordance was observed between trial enrollment assays (CTAs) and F1LCDx based on enrolled patients

– These findings demonstrate the clinical utility of the F1LCDx plasma assay as a non-invasive test to select *BRCA*-mutated mCRPC patients to receive the NIRA+AAP treatment

Please scan QR code

Poster

https://www.congresshub.com/Oncology/ESMO2024/Niraparib/Attard

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Acknowledgments

The authors thank the study patients and investigators for their participation. Shweta Pitre, M.Pharm, MPH CMPP (SIRO Clinpharm UK Limited) provided medical writing assistance and Jennifer Han, MS (Janssen Global Services, LLC) provided additional editorial support. Amit Kavle (SIRO Clinpharm Pvt. Ltd. India) provided graphic designing support.

Disclosures

G Attard: Honoraria-Janssen, Astellas Pharma; Consulting or Advisory Role- Janssen-Cilag, Veridex, Ventana Medical Systems, Astellas Pharma, Medivation, Novartis, Millennium, Abbott Laboratories, ESSA, Bayer, Pfizer, AZ, Ferring; Speakers' Bureau- Janssen,Astellas Pharma, Takeda, Sanofi, Ventana Medical Systems, Ipsen, AZ, Ferring; Research Funding-Janssen, Arno Therapeutics, Innocrin Pharma; Patents, Royalties, Other Intellectual Property- on The ICR rewards to inventors list of Abiraterone acetate; Travel, Accommodations, Expenses- Janssen, Astellas Pharma, Medivation, Ventana Medical Systems, Abbott Laboratories, Bayer, ESSA, Janssen, Astellas Pharma, Pfizer, Ferring; Other Relationship- Institute of Cancer Research.

Introduction

- Nearly one third of patients with metastatic castration-resistant prostate cancer (mCRPC) carry genetic mutations associated with the homologous recombination repair (*HRR*) pathway, leading to poor prognosis[1-3]
- Poly-ADP ribose polymerase inhibitors (PARPi) have demonstrated significant efficacy and clinical benefit in mCRPC patients with *HRR* alterations, particularly *BRCA1/2*-mutations (*BRCA+*)[4-7]
- The MAGNITUDE study demonstrated that the PARPi niraparib, in combination with abiraterone acetate plus prednisone (NIRA+AAP), significantly improved radiographic progression-free survival (rPFS) and other important clinical outcomes in *HRR+* mCRPC patients[7]
- NIRA+AAP is approved for patients with *BRCA+* mCRPC, as identified by approved companion diagnostic tests, FoundationOne®CDx (F1CDx) and FoundationOne® Liquid CDx (F1LCDx)

Objective

- To evaluate the clinical utility of a plasma assay F1LCDx in identifying *BRCA+* patients in the MAGNITUDE study

Methods

- The MAGNITUDE was a global, phase 3, double-blind, placebo (PBO)-controlled study (NCT03748641) evaluating NIRA+AAP versus PBO+AAP for the treatment of patients with mCRPC[7] (**Figure 1**)

Figure 1: MAGNITUDE Study Timeline

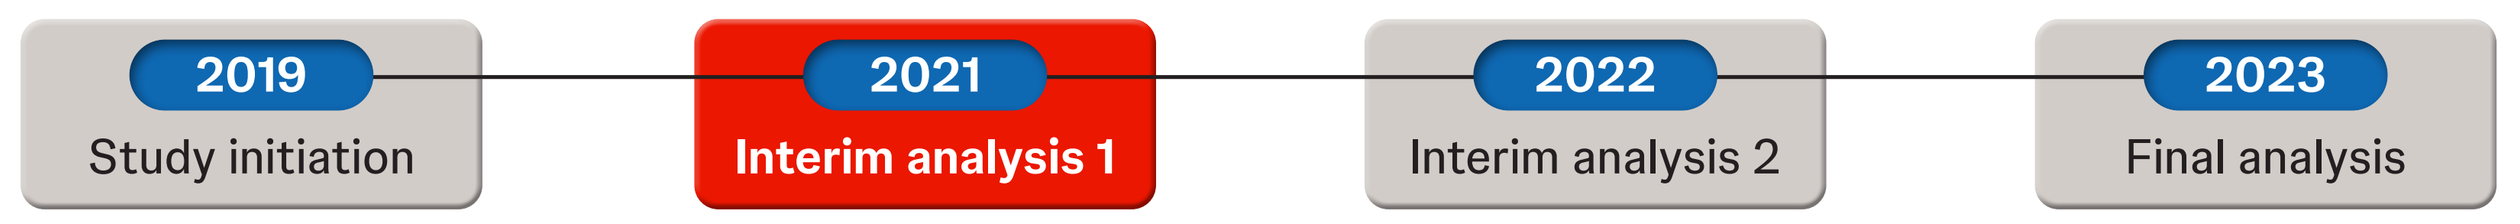

2019
Study initiation
2021
Interim analysis 1
2022
Interim analysis 2
2023
Final analysis

Study start: February 2019; clinical data cutoff for interim analysis 1 was October 8, 2021, for interim analysis 2 was June 17, 2022 and the final analysis was May 15, 2023.

- *HRR* and *BRCA* status was prospectively evaluated in the MAGNITUDE study using both tissue-based F1CDx assay and the plasma-based Exact Sciences Resolution homologous recombination deficiency (HRD)™ (Resolution HRD assay) clinical trial assays (CTA; **Figure 2**)

Figure 2: MAGNITUDE Study Design

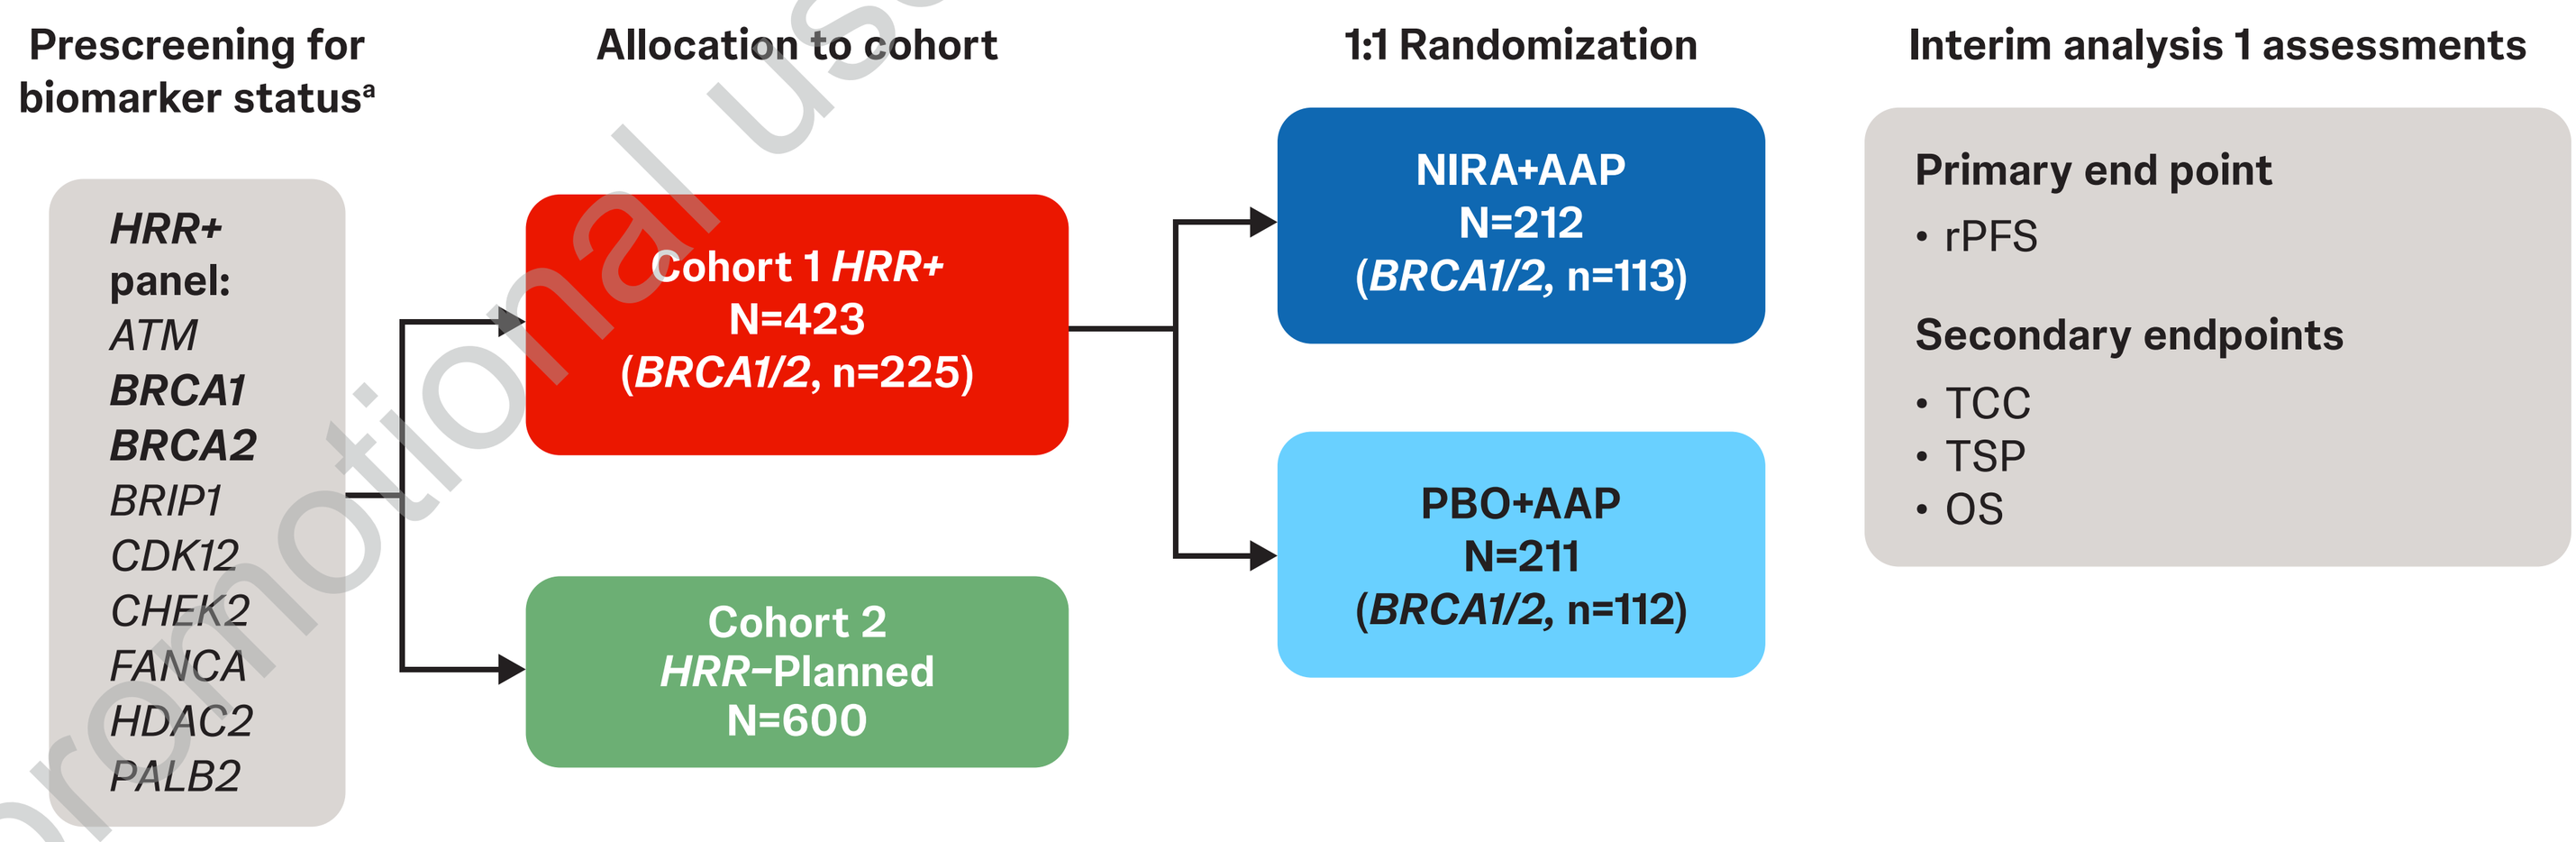

Prescreening for biomarker status[a]
Allocation to cohort
1:1 Randomization
Interim analysis 1 assessments
HRR+ panel: ATM BRCA1 BRCA2 BRIP1 CDK12 CHEK2 FANCA HDAC2 PALB2
Cohort 1 HRR+ N=423 (BRCA1/2, n=225)
Cohort 2 HRR–Planned N=600
NIRA+AAP N=212 (BRCA1/2, n=113)
PBO+AAP N=211 (BRCA1/2, n=112)
Primary end point
• rPFS
Secondary endpoints
• TCC
• TSP
• OS

[a]Patients were prospectively tested by plasma, tissue, and/or saliva/whole blood. Patients were required to provide both tissue and blood for prospective testing.
Tissue and plasma assays used included FoundationOne® CDx test (FoundationOne®CDx), Exact Sciences Resolution homologous recombination deficiency (HRD)™ (Resolution HRD assay) at central lab, or local lab biomarker test results demonstrating a pathogenic germline or somatic alteration outlined in the study protocol. The patients in the *HRR+* cohort harbored either pathogenic or likely pathogenic gene alterations in ≥1 of the following genes: *ATM, BRCA1, BRCA2, BRIP1, CDK12, CHEK2, FANCA, HDAC2,* or *PALB2*. The *HRR–* cohort included patients who had no detectable alterations in any of these genes or were undetectable.
AAP, abiraterone acetate with prednisone; *HRR*, homologous recombination repair; NIRA, niraparib; OS, overall survival; PBO, placebo; rPFS, radiographic progression-free survival; TCC, time to initiation of cytotoxic chemotherapy; TSP, time to symptomatic progression.

- Plasma testing using the liquid-based F1LCDx assay was performed retrospectively
- Concordance of *BRCA* status by F1LCDx with (a) CTAs and (b) tissue F1CDx was evaluated
- Clinical utility of F1LCDx was explored by comparing the primary endpoint of rPFS at the first interim analysis (median duration of follow-up: 18.6 months [range, 0.3–29.0 months]) between treatment arms in *BRCA+* patients identified by F1LCDx and enrolled by (a) CTAs and by (b) F1CDx in the MAGNITUDE study

Statistical analysis

- The Kaplan–Meier product limit method and a stratified Cox model were used to estimate rPFS and to obtain hazard ratios (HRs) with associated confidence intervals (CIs)

Results

Concordance

- Out of 473 patients from Cohort 1 (n=423) and Cohort 2 (n=50), 443 had plasma samples available for analysis. Of these 443 patients, 396 (89%) had evaluable F1LCDx results (**Figure 3**)

Figure 3: Patient Disposition

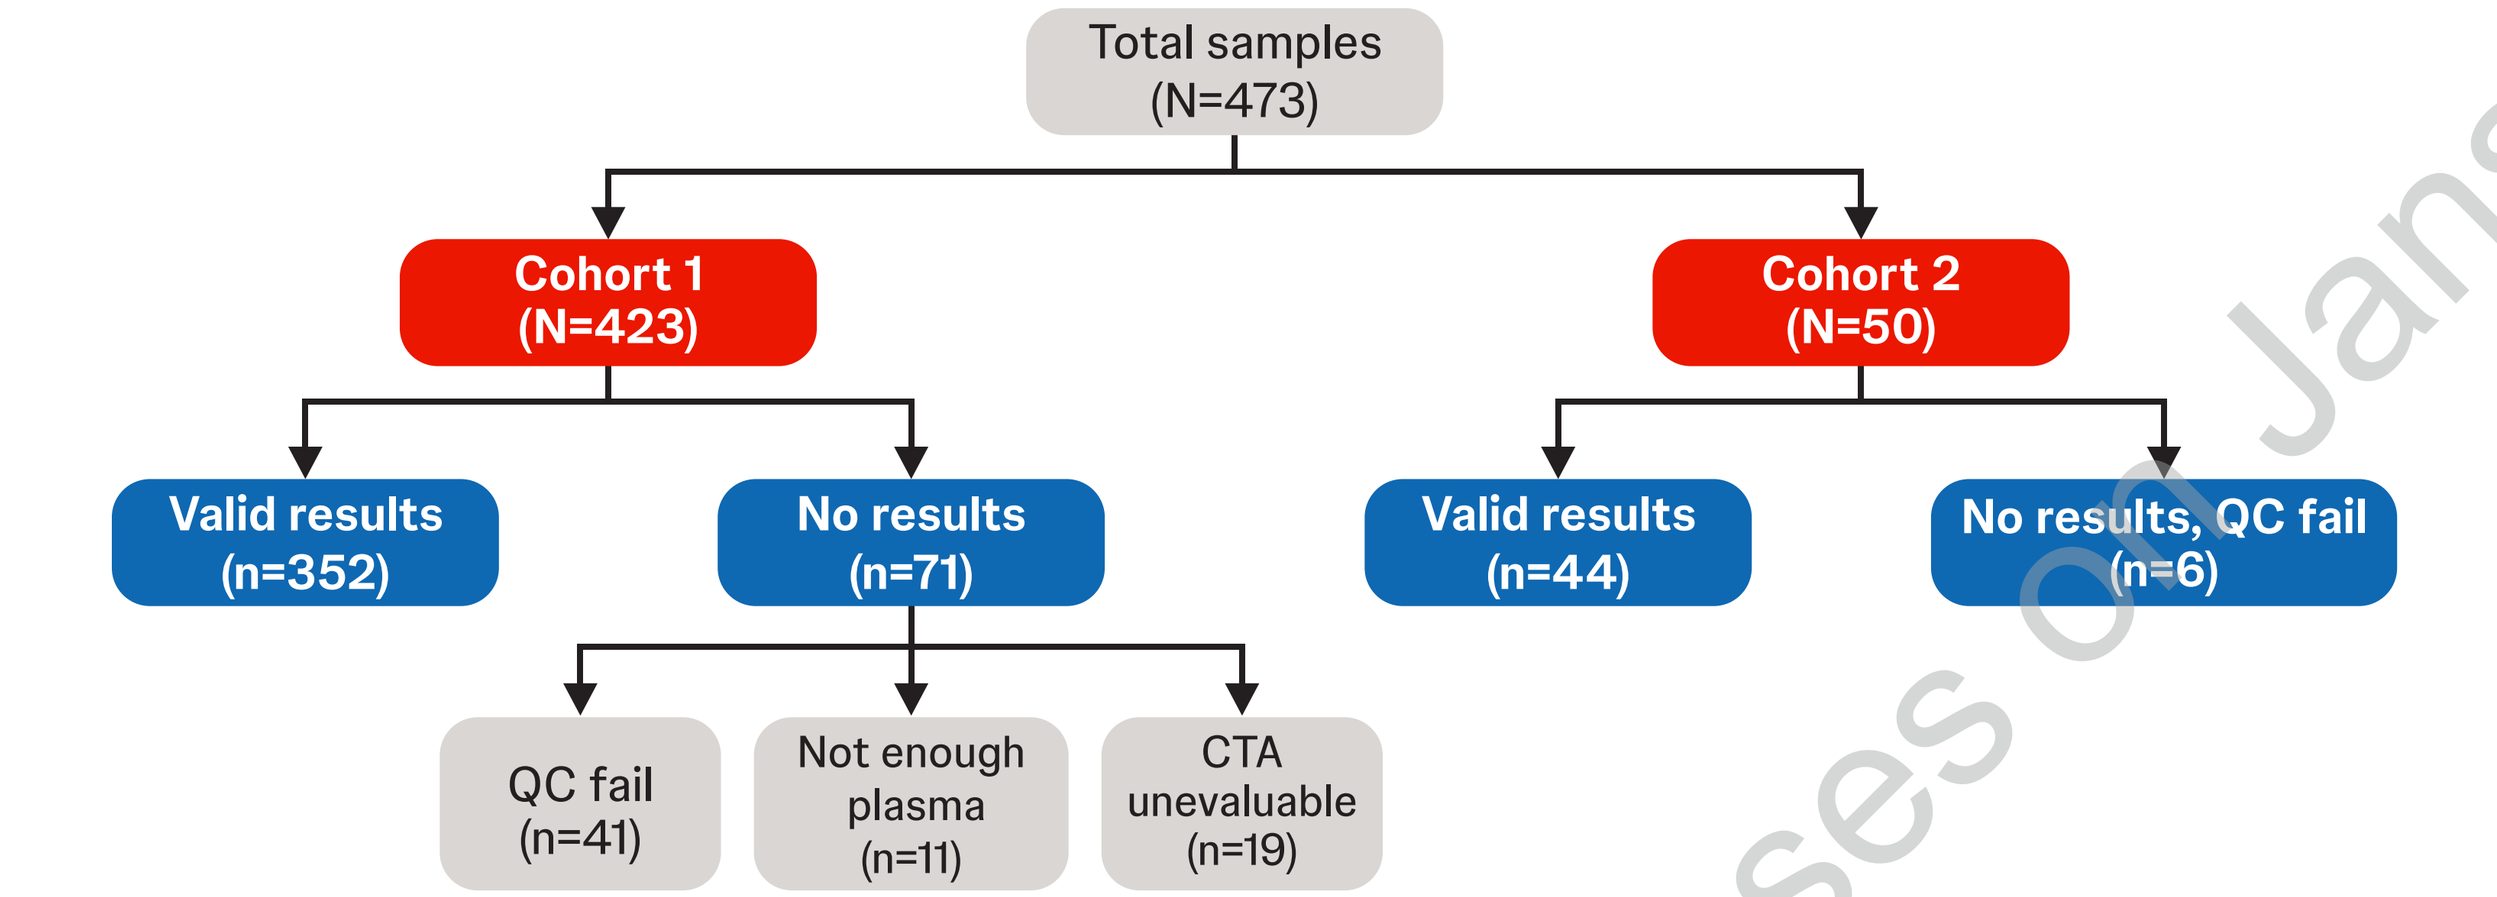

Total samples (N=473)
Cohort 1 (N=423)
Cohort 2 (N=50)
Valid results (n=352)
No results (n=71)
Valid results (n=44)
No results, QC fail (n=6)
QC fail (n=41)
Not enough plasma (n=11)
CTA unevaluable (n=19)

CTA, clinical trial assays; QC, quality control.

- Demographics and baseline clinical disease characteristics were broadly comparable across both, the CTA and F1LCDx assay groups (**Table 1**)

Table 1: Baseline Characteristics for CTA+ and F1CDx+ for *BRCA1/2* Patients

	CTA+ (N=216)	F1LCDx+ (N=142)
Age, years	68.17 (8.97)	67.91 (8.64)
Time from initial diagnosis to randomization, years	3.65 (3.62)	3.40 (3.67)
Time from metastatic diagnosis to first dose, years	0.41 (0.43)	0.41 (0.44)
Gleason total score 7 composition, n (%)	n=45	n=27
3 + 4	15 (6.9)	8 (5.6)
4 + 3	29 (13.4)	18 (12.7)
Unknown	1 (0.5)	1 (0.7)
ECOG performance status score, n (%)		
0	145 (67.1)	96 (67.6)
1	71 (32.9)	46 (32.4)
Baseline pain score (per BPI-SF item), n (%)	1.22 (1.8)	1.25 (1.7)
0	111 (51.4)	68 (47.9)
1 to 3	84 (38.9)	61 (43.0)
>3	20 (9.3)	12 (8.5)
Unknown	1 (0.5)	1 (0.7)
PSA at initial diagnosis (μg/L)	234.16 (636.64)	253.80 (697.52)

Data are shown as mean (SD), except mentioned otherwise. P-values compare CTA+ versus F1LCDx+ patients.
BPI-SF, Brief pain inventory- short form; CTA, clinical trial assays; ECOG, Eastern Cooperative Oncology Group; F1LCDx, FoundationOne®LiquidCDx; PSA, prostate-specific antigen.

- In the MAGNITUDE study, a total of 423 *HRR+* patients were enrolled in Cohort 1 using mainly CTAs (F1CDx tissue and Resolution HRD plasma assays), including 225 patients with *BRCA+* mutations (**Table 2**)
- Positive concordant results between the CTAs and F1LCDx assays were observed in 136 patients. Similarly, 109 patients showed positive concordance results between F1LCDx and F1CDx assays (**Table 3**)

Table 2: MAGNITUDE Enrollment in Cohort 1 with F1CDx Tissue Assay and Resolution HRD Assay

		F1CDx (Tissue)				
		BRCA1/2	Other *HRR*	Negative	No Result	Total
Resolution HRD assay (Plasma)	*BRCA*	96	5	12	37	150
	Other *HRR*	6	52	26	43	127
	Negative	56	68	0	3[a]	127
	No Result	4	4	0	11[b]	19
	Total	162	129	38	94	423

[a]Enrolled by local tissue assays through Foundation Medicine, no tissue collected for tissue central confirmation.
[b]Enrolled in China (by local AmoyDx test) and all of them are *BRCA2* positive.
F1CDx, FoundationOne®CDx; *HRR*, homologous recombination repair.

- The positive percent agreement (PPA) of 73.5% for F1LCDx with CTA as the reference indicates a moderate level of sensitivity for detecting positive results, while the negative percent agreement (NPA) of 97.2% for F1LCDx indicates a high level of specificity for confirming negative results (**Table 3**)
- The PPA of 80.7% for F1LCDx with F1CDx as the reference indicates a moderate level of sensitivity for detecting positive results, while the NPA of 96.0% for F1LCDx indicates a high level of specificity for confirming negative results (**Table 3**)
- Among samples with valid results from the F1LCDx and CTA assays, the prevalence-adjusted positive predictive value for F1LCDx vs CTA was 72.8% (95% CI: 58.6–89.3) and negative predictive value for F1LCDx vs CTA was 97.2% (95% CI: 96.6–97.9; **Table 3**)

Table 3: *BRCA* Concordance Between the F1LCDx and CTA (F1CDx+ Resolution HRD Assay) and F1CDx Tissue Assays

	CTA			F1CDx[a]		
F1LCDx	Positive	Negative	No Result	Positive	Negative	No Result
Positive (n=142)	136	6	0	109	7	26
Negative (n=254)	49	205	0	26	170	58
No Result (n=77)	31	35	11	27	27	23
Total (n=473)	216	246	11	162	204	107
PPA (95% CI)	**73.5% (66.7–79.3)**			**80.7% (73.3–86.5)**		
NPA (95% CI)	**97.2% (93.9–98.7)**			**96.0% (92.1–98.1)**		
Adjusted PPV (95% CI)	72.8% (58.6–89.3)			69.6% (56.2–87.8)		
Adjusted NPV (95% CI)	97.2% (96.6–97.9)			97.8% (97.0–98.5)		

[a]2 out 3 patients enrolled by Foundation Medicine local tissue assays are *BRCA* positives by these local assays. These local results were not used in defining the *BRCA* status by CTA.
CTA, clinical trial assays; CI, confidence interval; F1CDx, FoundationOne®CDx; F1LCDx, FoundationOne®LiquidCDx; NPA, negative percent agreement; NPV, negative predictive value; PPA, positive percent agreement; PPV, positive predictive value.

Clinical Utility

- Median rPFS in all *BRCA+* patients detected by CTAs was 16.6 (95% CI: 14.4–NE) months in the NIRA+AAP group and 10.9 (95% CI: 8.3–13.8) months in the PBO+AAP group (HR: 0.54 [95% CI: 0.36–0.81]; p=0.002; **Figure 4**)
- Within the CTA *BRCA+* group, median rPFS in all *BRCA+* patients detected by F1LCDx was 18.4 (95% CI: 13.8–NE) months in the NIRA+AAP group and 9.0 (95% CI: 8.2–16.4) months in the PBO+AAP group (HR: 0.49 [95% CI: 0.29–0.81]; p=0.005; **Figure 4**)
- Median rPFS in all *BRCA+* patients detected by F1CDx was 18.4 (95% CI: 16.1–NE) months in the NIRA+AAP group and 10.9 (95% CI: 8.4–13.8) months in the PBO+AAP group (HR: 0.46; [95% CI: 0.29–0.73]; p=0.001; **Figure 4**)
- Within the F1CDx *BRCA+* group, median rPFS in F1LCDx *BRCA+* patients was 18.4 (95% CI: 13.4–NE) months in the NIRA+AAP group and 8.4 (95% CI: 7.3–13.8) months in the PBO+AAP group (HR: 0.42 [95% CI: 0.24–0.74]; p=0.002; **Figure 4**)
- Within Cohort 1 of patients enrolled by CTAs, the rPFS following NIRA+AAP treatment was comparable in *BRCA+* patients identified by CTAs and by F1LCDx (**Figure 4**)

Figure 4: Efficacy by Assays

Subgroup	Patients (events) NIRA+AAP	Patients (events) PBO+AAP	Median rPFS (months) NIRA+AAP	Median rPFS (months) PBO+AAP	Hazard ratio (95% CI)
Total CTA+	106 (44)	110 (64)	16.6 (14.4–NE)	10.9 (8.3–13.8)	0.54 (0.36–0.81)
F1LCDx+\|CTA+	64 (25)	72 (44)	18.4 (13.8–NE)	9.0 (8.2–16.4)	0.49 (0.29–0.81)
F1LCDx–\|CTA+	29 (11)	20 (11)	16.7 (15.0–NE)	8.4 (8.3–NE)	0.39 (0.15–1.03)
F1LCDx unevaluable\|CTA+	13 (8)	18 (9)	13.9 (11.1–NE)	11.2 (8.0–NE)	0.75 (0.25–2.23)
Total F1CDx+	76 (30)	86 (51)	18.4 (16.1–NE)	10.9 (8.4–13.8)	0.46 (0.29–0.73)
F1LCDx+\|F1CDx+	49 (20)	60 (38)	18.4 (13.4–NE)	8.4 (7.3–13.8)	0.42 (0.24–0.74)
F1LCDx–\|F1CDx+	16 (3)	10 (5)	NE (16.1–NE)	8.4 (8.4–NE)	0.12 (0.02–0.68)
F1LCDx unevaluable\|F1LCDx+	11 (7)	16 (8)	13.1 (10.9–NE)	11.2 (7.6–NE)	1.04 (0.32–3.36)

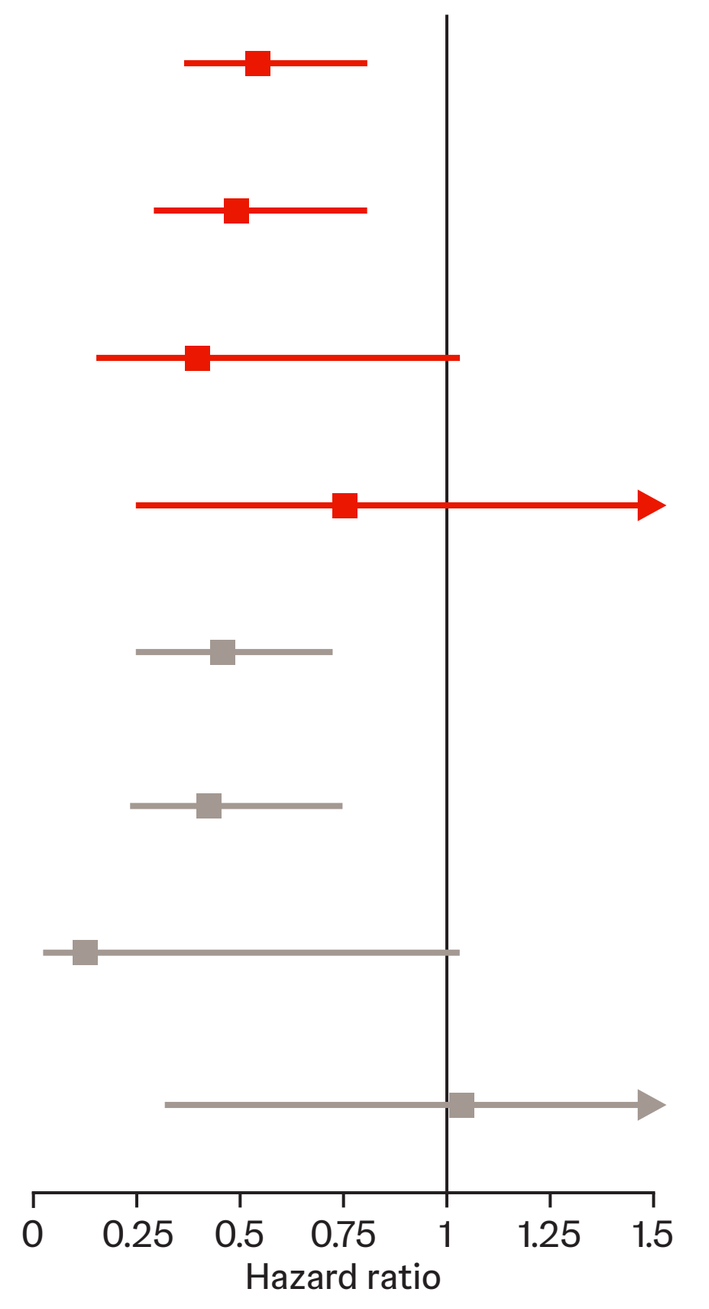

0
0.25
0.5
0.75
1
1.25
1.5
Hazard ratio

rPFS is expressed as median (95% CI). AAP, abiraterone acetate plus prednisone; CTA, clinical trial assays; F1CDx, FoundationOne®CDx tissue assay; F1LCDx, FoundationOne®Liquid CDx; NE; not evaluable; NIRA, niraparib; PBO, placebo; rPFS, radiographic progression-free survival.

Limitations

- The analysis did not assess homozygous copy number loss of *BRCA* genes in F1LCDx, which might require a higher ctDNA tumor fraction for detection and could influence the observed results[8-10]
- Homozygous *BRCA* gene loss is associated with significant benefit from PARPi in mCRPC,[10-11] due to the inability to form reversion mutations.[12] However, due to the small number of events in the F1LCDx–/F1CDx+ subgroup which includes homozygous *BRCA* loss patients by F1CDx, this hypothesis could not be further investigated

References

1. Leith A, et al. *Future Oncol.* 2022;18:937-951.
2. Castro E, et al. *J Clin Oncol.* 2019;20;37:490-503.
3. Abida W, et al. *JCO Precis. Oncol.* 2017;2017:PO.17.00029.
4. Mateo J. et al. *Lancet Oncol.* 2020;21:162-174.
5. de Bono J, et al. *Lancet Oncol.* 2021,22:1250-1264.
6. Abida W, et al. *J Clin Oncol.* 2020;38:3763-3772.
7. Chi K, et al. *J Clin Oncol.* 2023;41:3339-3351.
8. Woodhouse R, et al. *Plos One.* 2020, 15:e0237802.
9. Chi K, et al. *Clin Cancer Res.* 2023, 29: 81–91.
10. Triner D, et al. *ESMO Open.* 2024, (in press).
11. Mateo J, et al. *J Clin Oncol.* 2024, 42:571-583.
12. Loehr A, et al. *Eur Urol.* 2023, 83:200-209.

Prostate Cancer

This material is distributed for scientific purposes on Janssen Science, and is not for promotional use

Presented by Dr. Attard at the European Society of Medical Oncology (ESMO) Congress 2024, September 13-17 | Barcelona, Spain