1980P

Association of PD-L1 Expression With Clinical Response to TAR-200 in the Phase 2b SunRISe-1 Trial

Evanguelos Xylinas[1], Christopher M Pieczonka[2], Joseph M Jacob[3], Andrea Necchi[4], Siamak Daneshmand[5], Giuseppe Simone[6], Karel Decaestecker[7], Girish S Kulkarni[8], Martin Bögemann[9], Félix Guerrero Ramos[10], Justin Callaway[11], Neil Beeharry[11], Jiarui Zhang[11], Shalaka Hampras[12], Hussein Sweiti[11], Katharine Stromberg[12], Jason Martin[13], Abhijit Shukla[14], Patricia Raciti[11], Michiel S van der Heijden[15]

[1]Department of Urology, Bichat-Claude Bernard Hospital, Assistance Publique-Hôpitaux de Paris, Université de Paris Cité, Paris, France; [2]Associated Medical Professionals of New York, Syracuse, NY, USA; [3]Department of Urology, Upstate Medical University, Syracuse, NY, USA; [4]IRCCS San Raffaele Hospital, Vita-Salute San Raffaele University, Milan, Italy; [5]Department of Urology, University of Southern California Norris Comprehensive Cancer Center, Los Angeles, CA, USA; [6]Department of Urology, Regina Elena National Cancer Institute, Rome, Italy; [7]Department of Urology, AZ Maria Middelares, Ghent, Belgium; [8]Division of Urology, Department of Surgery, University Health Network, University of Toronto, Toronto, ON, Canada; [9]Department of Urology, Muenster University Hospital, Muenster, Germany; [10]12 de Octubre University Hospital, Madrid, Spain; [11]Janssen Research & Development, Spring House, PA, USA; [12]Clinical Oncology, Janssen Research & Development, Raritan, NJ, USA; [13]Janssen Research & Development, High Wycombe, UK; [14]Clinical Oncology, Janssen Research & Development, Lexington, MA, USA; [15]Department of Medical Oncology, Netherlands Cancer Institute, Amsterdam, Netherlands

Key Takeaway

In this exploratory analysis, TAR-200 monotherapy showed a high CR rate in patients with BCG-unresponsive HR NMIBC CIS irrespective of PD-L1 and TMB status

Conclusions

Across subgroups of patients with BCG-unresponsive HR NMIBC CIS based on PD-L1 status, TAR-200 monotherapy showed a high CR rate (85%-91%)

A high CR rate (94%) was observed in the subgroup of patients with TMB low status who received TAR-200 monotherapy; the sample size in the TMB high subgroup was very small (n=4)

All patients with available MSI scores had MSS tumors; the CR rate among patients with MSS tumors was also high (86%)

Scan the QR code

Poster

https://www.congresshub.com/Oncology/ESMO2024/TAR-200/Xylinas

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Acknowledgments
The authors would like to thank the patients who participated in the study, their families, and the investigators and clinical research staff from the study centers. This study was funded by Janssen Research & Development. Writing assistance was provided by Jennifer Venzie, PhD, of Parexel, and was funded by Janssen Global Services, LLC. Data analysis assistance was provided by Rui Hong, PhD, of Janssen Research & Development.

Disclosures
E Xylinas has received grants/research funding from Ferring; and consulting/advisory fees from Astellas, AstraZeneca, Boston Scientific, Bristol Myers Squibb, Ipsen, Janssen Oncology, and MSD.

Introduction

- Recurrent non–muscle-invasive bladder cancer (NMIBC) tumors after bacillus Calmette-Guérin (BCG) treatment are characterized by an immunosuppressive environment and increased programmed death ligand 1 (PD-L1) expression[1]
- TAR-200 is a novel gemcitabine intravesical system designed for sustained, local delivery of chemotherapy within the bladder[2,3]
 - TAR-200 is placed in the bladder in a brief in-office procedure
- SunRISe-1 is an ongoing phase 2b trial evaluating the efficacy and safety of TAR-200 in BCG-unresponsive patients with high-risk (HR) NMIBC who are ineligible for or decline radical cystectomy (RC) (Figure 1)
- We evaluated the association between PD-L1 expression, tumor mutational burden (TMB), and microsatellite instability (MSI) and response to TAR-200 monotherapy in Cohort 2 of SunRISe-1

Figure 1: SunRISe-1 Cohort 2 (NCT04640623) study design and biomarker measurement

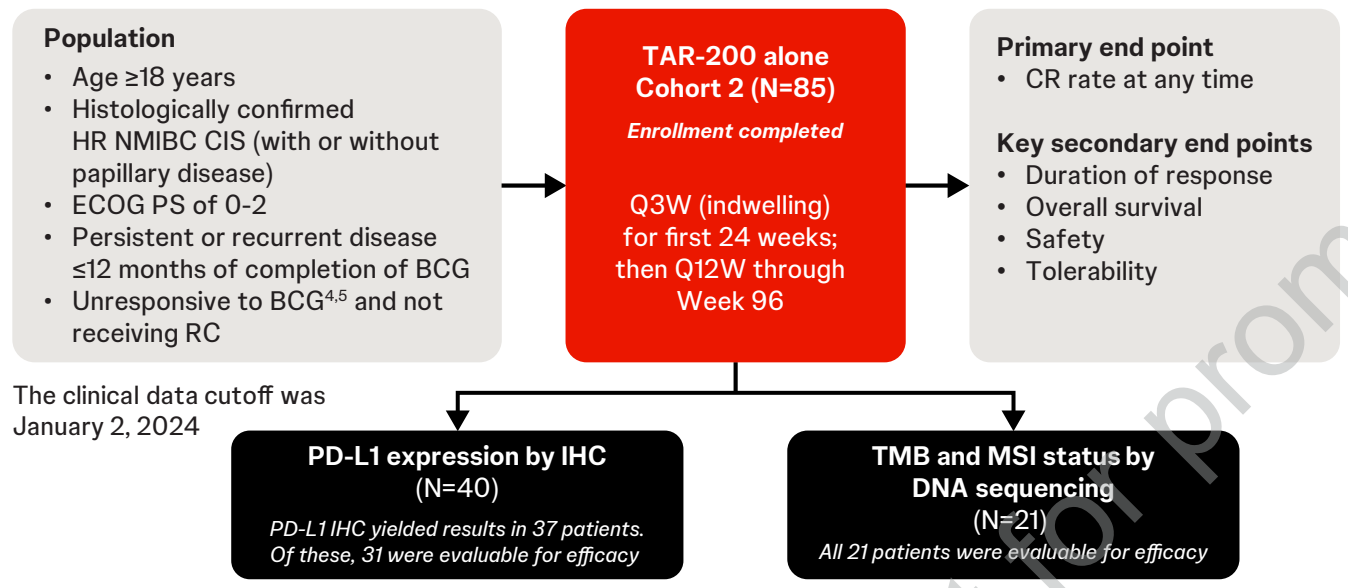
Population
Age ≥18 years
Histologically confirmed HR NMIBC CIS (with or without papillary disease)
ECOG PS of 0-2
Persistent or recurrent disease ≤12 months of completion of BCG
Unresponsive to BCG[4,5] and not receiving RC
TAR-200 alone Cohort 2 (N=85)
Enrollment completed
Q3W (indwelling) for first 24 weeks; then Q12W through Week 96
Primary end point
CR rate at any time
Key secondary end points
Duration of response
Overall survival
Safety
Tolerability
The clinical data cutoff was January 2, 2024
PD-L1 expression by IHC (N=40)
PD-L1 IHC yielded results in 37 patients. Of these, 31 were evaluable for efficacy
TMB and MSI status by DNA sequencing (N=21)
All 21 patients were evaluable for efficacy

CR, complete response; ECOG PS, Eastern Cooperative Oncology Group performance status; IHC, immunohistochemistry; Q3W, every 3 weeks; Q12W, every 12 weeks.

Methods

- Clinical outcomes in these analyses used the January 2, 2024, clinical data cutoff for SunRISe-1[6]
- Tumor tissue was collected during screening by transurethral resection of bladder tumor or bladder biopsy from patients with HR NMIBC carcinoma in situ (CIS), with or without papillary disease, enrolled in Cohort 2 of SunRISe-1
- PD-L1 was measured by IHC (22C3) and scored for combined positive score (CPS) with cutoffs of 1 and 10
- DNA isolation and sequencing using the Illumina TruSight Oncology 500 panel measured TMB and MSI scores
 - MSI high was defined as ≥20% of unstable microsatellite sites; microsatellite stable (MSS) was defined as <20%
- Fisher's exact test was used to determine p values in comparisons of biomarker subgroups

Results

Patients

- At a January 2, 2024, interim data cutoff, 85 patients had received TAR-200 monotherapy (Table 1)
- PD-L1 expression was measured in a subset of 40 patients in Cohort 2 with available tumor tissue, with 31 yielding PD-L1 results and being evaluable for efficacy (Table 2)
 - Of these, 11 (35%) had tumors with PD-L1 CPS ≥10 and 20 (65%) had tumors with PD-L1 CPS ≥1
- TMB and MSI were assessed in 21 patients using baseline DNA isolation and sequencing from available tumor tissue (Table 2)
 - 17 patents (81%) had TMB low tumors; 4 (19%) had TMB high
 - All 21 patients had MSS tumors (100%), with a median 1.6% of unstable microsatellite sites; none had MSI high tumors

Table 1: Patient characteristics in the TAR-200 monotherapy cohort of SunRISe-1

Characteristics	TAR-200 Monotherapy N=85[a]
Age, median (range), y	71 (40-88)
Male, n (%)	68 (80)
Race, n (%)	
White	62 (73)
Asian	8 (9)
Black/African American	2 (2)
Not reported/Unknown	13 (15)
Nicotine use, n (%)	
Current	8 (9)
Former	49 (58)
Never	28 (33)
ECOG PS 0, n (%)	78 (92)
Tumor stage, n (%)	
CIS only	57 (67)
CIS + papillary disease	28 (33)
Total prior BCG doses, median (range), n	12 (7-42)
Time from last BCG to CIS diagnosis, median (range), mo	3 (0-22)[b]
Reason for not receiving RC, n (%)	
Declined	82 (96)
Ineligible	3 (4)

[a]Patient characteristics are shown for all patients who received at least 1 dose of TAR-200 in the full analysis set (N=85). [b]1 patient had 22.4 months from last BCG dose to CIS diagnosis (protocol deviation); all other patients had ≤12 months from last BCG dose to CIS diagnosis (per protocol).

Table 2: Biomarker status in patients with HR NMIBC treated with TAR-200 monotherapy in SunRISe-1

Biomarker Status, n (%)	TAR-200 Monotherapy N=85
PD-L1	**N=31[a]**
CPS ≥10	11 (35)
CPS <10	20 (65)
CPS ≥1	20 (65)
CPS <1	11 (35)
TMB	**N=21[a]**
TMB high	4 (19)
TMB low	17 (81)
MSI	**N=21[a]**
MSS	21 (100)
MSI high	0

[a]Percentages are based on number of patients with available data.

Efficacy in Biomarker Subgroups

- At the data cutoff used for these analyses, 58 patients were efficacy evaluable; TAR-200 monotherapy showed a CR rate at any time of 83% (95% CI, 71-91)[6]
- Among 31 patients with available PD-L1 scores who were treated with TAR-200 monotherapy and were efficacy evaluable, the CR rate was 87% (27/31)
 - CR rates of 91% and 85% were observed in the CPS ≥10 and CPS <10 subgroups, respectively (Figure 2A)
 - CR rates of 85% and 91% were observed in the CPS ≥1 and CPS <1 subgroups, respectively (Figure 2B)
- In 21 patients with available TMB data who were treated with TAR-200 monotherapy and were efficacy evaluable, the CR rate was 86% (18/21)
 - CR rates of 94% and 50% were observed in the TMB low and TMB high subgroups, respectively (Figure 2C)
- In 21 patients with available MSI scores who were treated with TAR-200 monotherapy and were efficacy evaluable, the CR rate was 86% (18/21) (95% CI, 64-97)
- Across biomarker subgroups analyzed, no statistically significant differences in CR rates were observed

Figure 2: CR rate in subgroups of patients by biomarker status

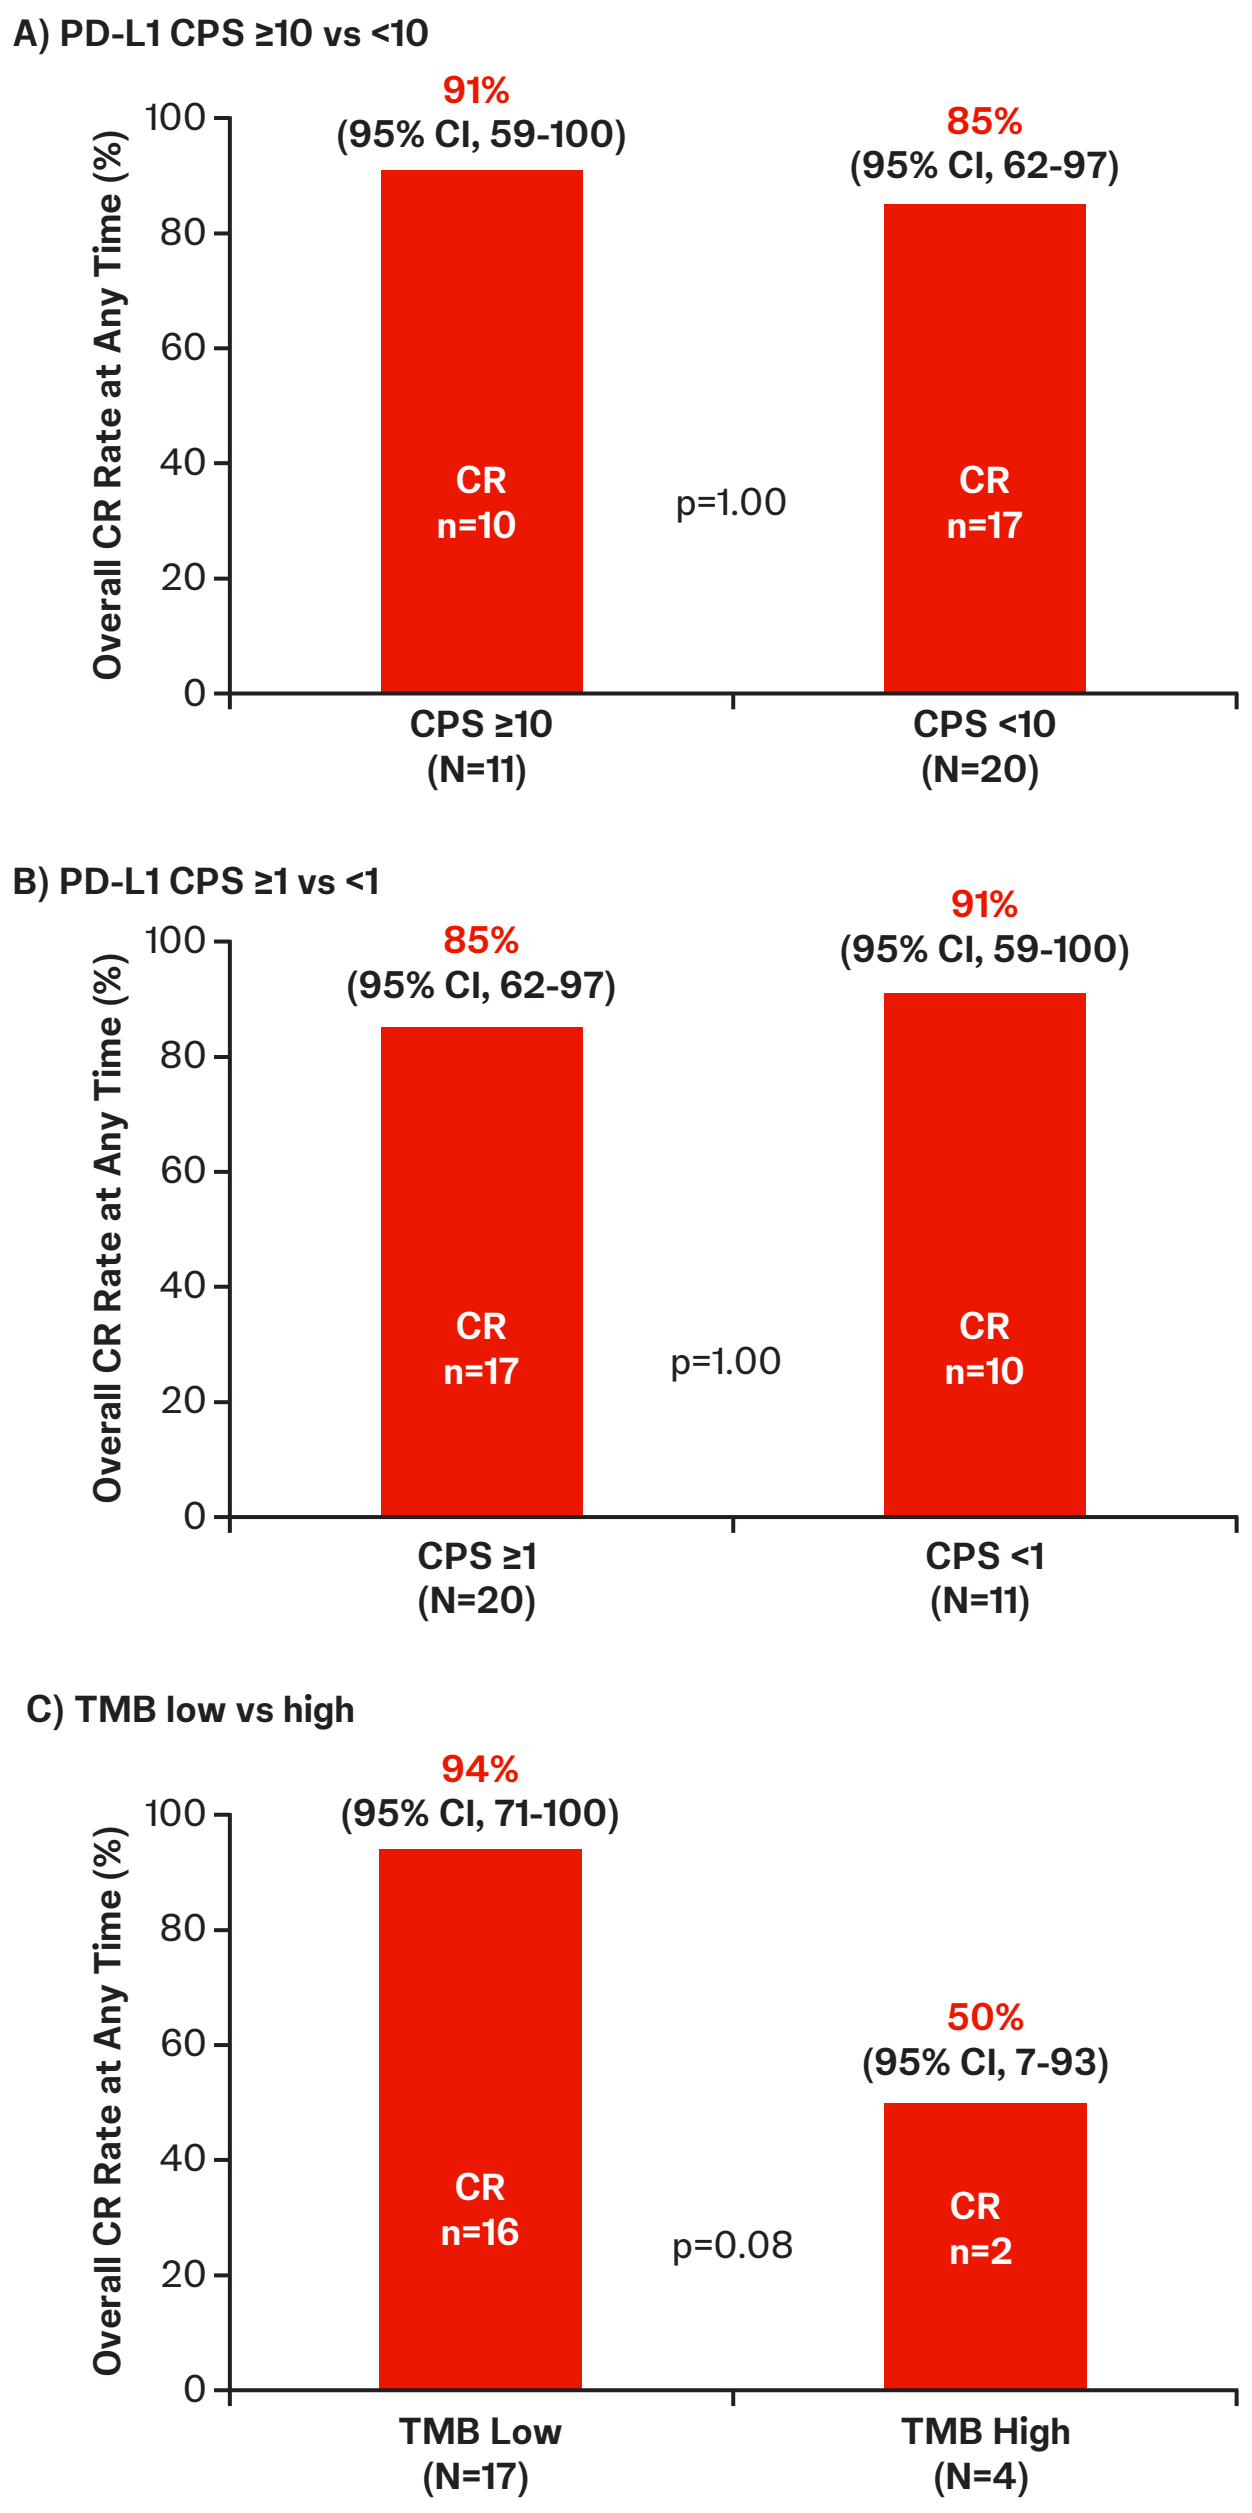
A) PD-L1 CPS ≥10 vs <10
Overall CR Rate at Any Time (%)
0
20
40
60
80
100
91%
(95% CI, 59-100)
CR
n=10
p=1.00
85%
(95% CI, 62-97)
CR
n=17
CPS ≥10
(N=11)
CPS <10
(N=20)
B) PD-L1 CPS ≥1 vs <1
85%
(95% CI, 62-97)
CR
n=17
p=1.00
91%
(95% CI, 59-100)
CR
n=10
CPS ≥1
(N=20)
CPS <1
(N=11)
C) TMB low vs high
94%
(95% CI, 71-100)
CR
n=16
p=0.08
50%
(95% CI, 7-93)
CR
n=2
TMB Low
(N=17)
TMB High
(N=4)

References
1. Hashizume A, et al. *Oncotarget.* 2018;9:34066-34078. 2. Daneshmand S, et al. *Urol Oncol.* 2022;40:344.e1-344.e9. 3. Tyson MD, et al. *J Urol.* 2023:209:890-900. 4. Lerner SP, et al. *Urol Oncol.* 2009;27:155-159. 5. US Food and Drug Administration. Available at: https://www.fda.gov/media/101468/download. 6. Jacob JM, et al. Presented at 119th AUA Annual Meeting, May 3-6, 2024, San Antonio, TX, USA.

Urothelial Cancer

Presented by E Xylinas at the European Society for Medical Oncology (ESMO) Congress 2024; September 13-17, 2024; Barcelona, Spain

This material is distributed for scientific purposes on Janssen Science, and is not for promotional use